1594

A fast and sensitive workflow to screen therapy-relevant mutations in circulating tumor cells and quantification of cancer-associated exosomal mRNA in plasma of cancer patients

DO NOT POST

Martin H.D. Neumann, Nina Kessler, Sebastian Bender, Thomas Krahn, Thomas Schlange

Bayer AG, Pharmaceutical Division, Wuppertal/Berlin, Germany

INTRODUCTION

- The tests analyzing rare circulating cells or nucleic acids in blood are considered liquid biopsies.
- Liquid biopsies offer a simple, fast, and cost-efficient way to monitor the status of a disease or the efficacy of a treatment at any time point with acceptable risk and burden for the patient.
- The detection requires robust, highly sensitive technologies like droplet digital PCR (ddPCR) as the volumes of available samples can be limited and rare cells as well as circulating nucleic acids are often present in low abundance.
- Here, we present a novel workflow combining effective enrichment of rare cells with the microfluidic filtration device Parsortix™ and the detection of point mutations with ddPCR (**Figure 1**). The approach enables the analysis of therapy-relevant point mutations and the quantification of circulating tumor cells (CTCs) as well as extracellular vesicle-derived mRNA (EVmRNA).

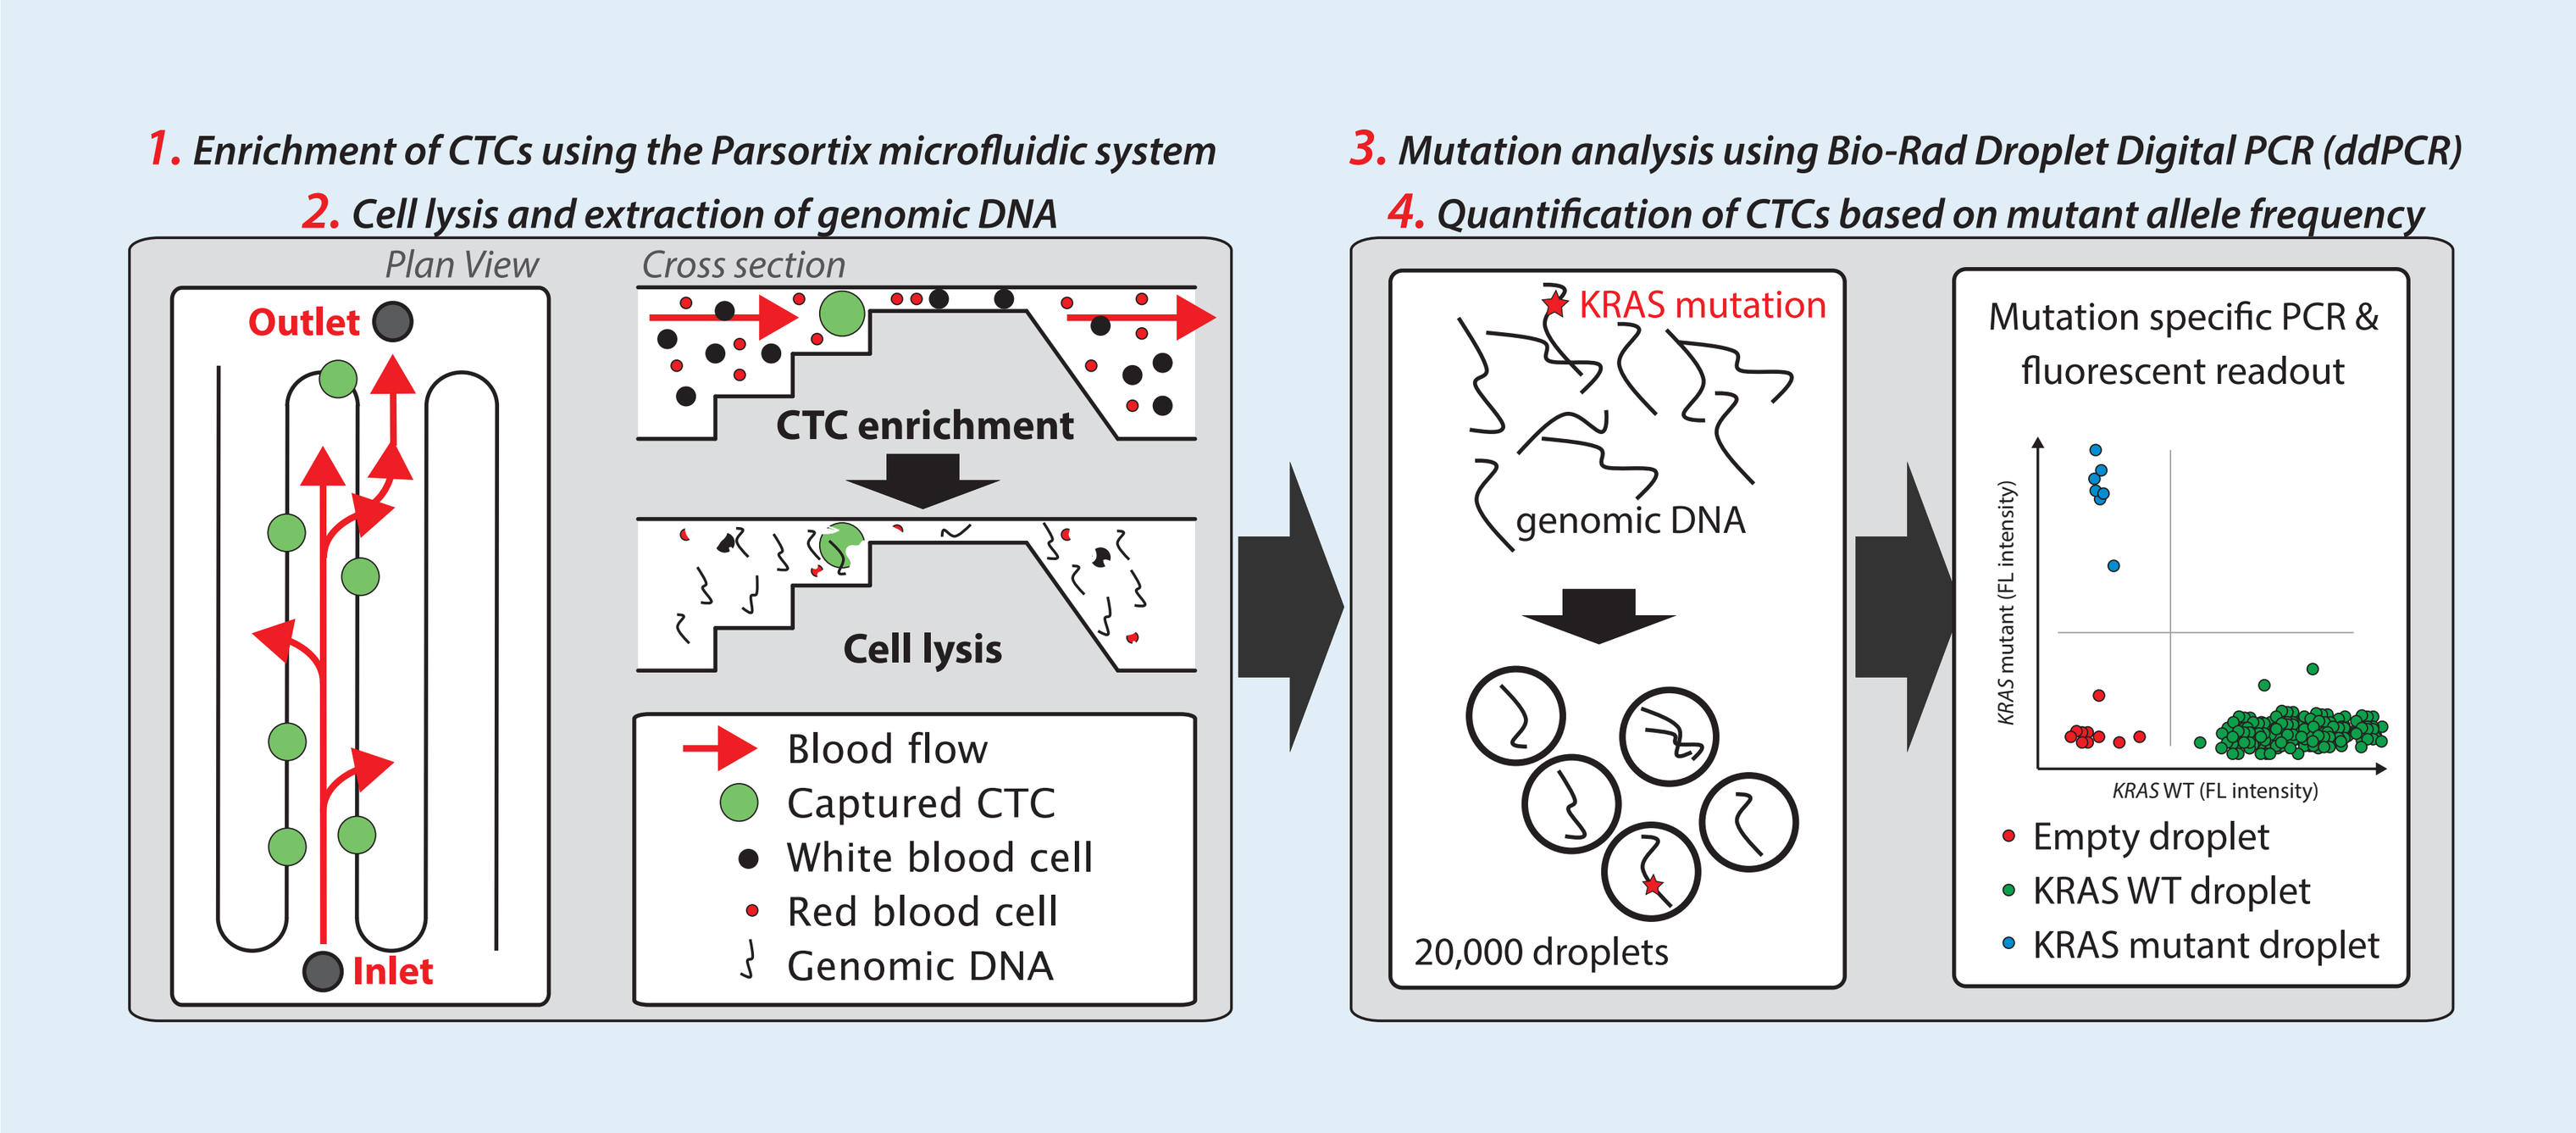
1. Enrichment of CTCs using the Parsortix microfluidic system
2. Cell lysis and extraction of genomic DNA
3. Mutation analysis using Bio-Rad Droplet Digital PCR (ddPCR)
4. Quantification of CTCs based on mutant allele frequency
Plan View
Cross section
Outlet
Inlet
CTC enrichment
Cell lysis
Blood flow
Captured CTC
White blood cell
Red blood cell
Genomic DNA
KRAS mutation
genomic DNA
20,000 droplets
Mutation specific PCR & fluorescent readout
KRAS mutant (FL intensity)
KRAS WT (FL intensity)
Empty droplet
KRAS WT droplet
KRAS mutant droplet

Figure 1. Workflow from a liquid biopsy to quantification of rare cells or nucleic acids. A simple, fast and cost-efficient way to analyze circulating tumor cells (CTCs) or nucleic acids in blood or other biofluids.

CONCLUSIONS

- As a screening approach, ddPCR allows the verification of therapy-relevant point mutations in CTCs. The sensitivity of point mutation determination by ddPCR is near to single cell level and in combination with the CTC counting, genetic information and CTC counts can be combined.
- Extracellular vesicles and especially exosomes, transporting small amounts of RNA, proteins and other factors, serve as a biomarker platform.
- Here, we show that EVmRNA can reflect treatment effect by showing an increase or decrease of target gene expression detectable by ddPCR. Moreover, EVmRNA may reflect the expression profile of cancer cells, but this is dependent on the target. EVmRNA could also be successfully detected and quantified in human plasma.
- A larger cohort and a deeper investigation on cell-based assays will follow to support these findings.

ACKNOWLEDGEMENTS

This work was supported by the Innovative Medicines Initiative Joint Undertaking under Grant Agreement n° 115749, resources of which are composed of financial contribution from the European Union's Seventh Framework Programme (FP7/2007-2013) and EFPIA. Samples from patients and healthy volunteers, respectively, were collected under signed informed consent. Aurexel Life Sciences Ltd. (www.aurexel.com) is thanked for editorial assistance in the preparation of this poster, funded by Bayer AG.

Poster presented at the AACR Annual Meeting, April 14-18, 2018, Chicago, IL, USA

RESULTS

A fast approach for screening point mutations in rare cells

- The novel workflow enabled the quantification of low numbers of circulating tumor cells (CTC) and their analysis for therapy-relevant point mutations (**Figure 2**).
- The *KRAS* G12V and *TP53* R158L mutations were detected at an expected range (**Figure 2B**), demonstrating that in-cassette staining, cell quantification and mutation analysis are applicable methods in the screening approach.

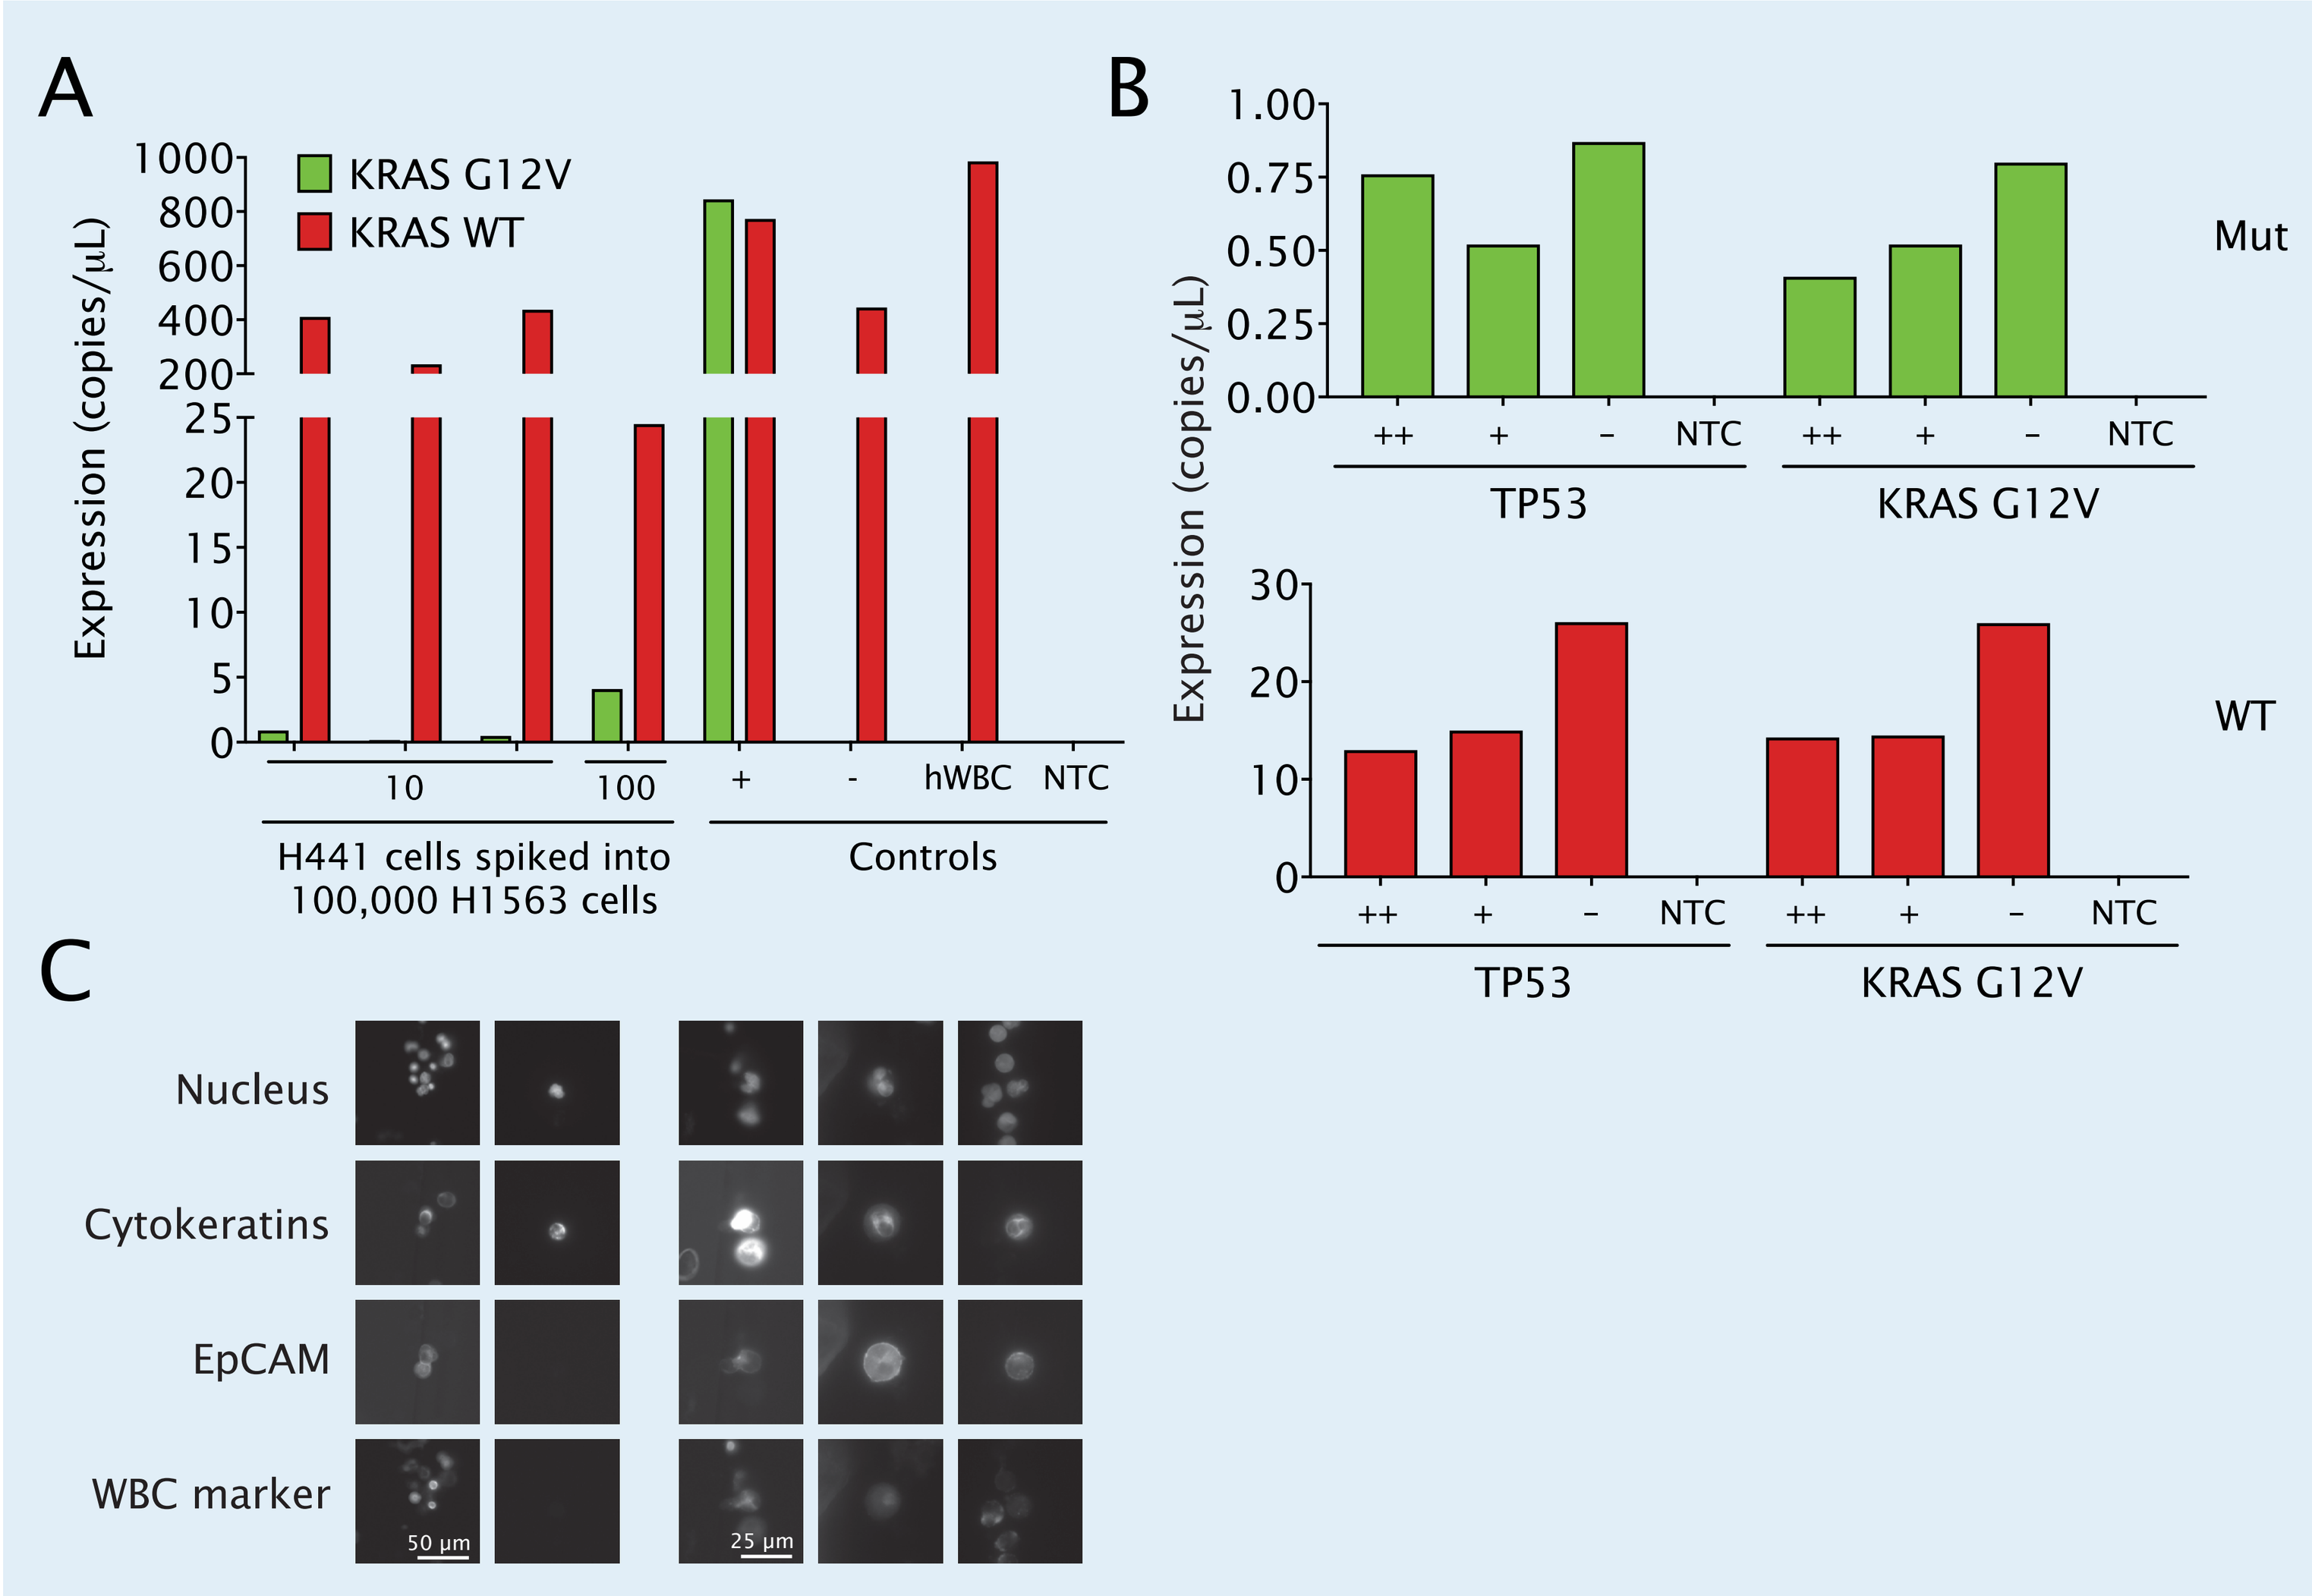
A
KRAS G12V
KRAS WT
Expression (copies/µL)
H441 cells spiked into 100,000 H1563 cells
Controls
10
100
+
-
hWBC
NTC
B
Expression (copies/µL)
Mut
WT
++
+
-
NTC
TP53
KRAS G12V
C
Nucleus
Cytokeratins
EpCAM
WBC marker
50 µm
25 µm

Figure 2. Detection of point mutations in cancer cells after rare cell enrichment. Non-small cell lung cancer (NSCLC) cells NCI-H441 (H441) carrying the heterozygous *KRAS* G12V and *TP53* R158L point mutations were added into a suspension of NCI-H1563 (H1563) cells with wild-type (WT) *KRAS* and *TP53*, or into K_2EDTA plasma samples pooled from 3 healthy donors (hBWC). The mixtures were processed using the Parsortix™ microfluidic device to enrich cell factions by size and deformability as described in Figure 1. Cells were harvested or lysed directly in the Parsortix cassette with or without in-cassette staining, followed by preparation of genomic DNA (gDNA).

A) ddPCR detection of *KRAS* WT and G12V mutation without in-cassette staining. 10 (three replicates) or 100 H441 cells were spiked into 100,000 H1563 cells. +, positive control, 100,000 H441 cells; -, negative control, 100,000 H1563; hWBC, healthy white blood cells; NTC, no template control. 1 copies/µL is equivalent to 20 haploid genome equivalents. We detected 0.1-0.9 copies/µL , which is equivalent to 1.5 – 13.5 H441 cells heterozygous for *KRAS* G12V.

B) For testing the quantitation limit of point mutations after Parsortix™ enrichment with and without in-cassette staining, 10 H441 cells were spiked into a suspension of 10,000 H1563 cells with a final volume of 2 mL PBS. Detection of 10 H441 cells either with (+) or without (-) in-cassette staining was compared to 10 H441 cells spiked into 4 mL of whole blood with in-cassette staining (++). The cell suspension was directed to in-cassette lysis followed by gDNA extraction and mutation analysis. We detected 0.5 – 0.8 copies/µL of mutated targets, which is equivalent to 7.5–12 haploid genome equivalents, i.e. 7.5 -12 heterozygous mutated cells. NTC, no template control; Mut, mutated, WT, wild-type.

C) Representative images of a mixture of H441 and H1563 cells with WBC background in the Parsortix™ cassette after staining for nucleus, cytokeratins, epithelial cell adhesion molecule (EpCAM) and the WBC-marker CD45 at two different magnifications.

Extracellular vesicle-derived mRNA (EVmRNA) reflects treatment response and gene expression in cancer cells

- Extracellular vesicles (EV) are actively secreted by cells, including cancer cells, and their cargo contains, amongst other, mRNA. To determine if EVmRNA can be used to monitor pharmacological effects, the amount of cancer-associated EVmRNA secreted by the cells after cisplatin treatment was quantified.
- Quantification of vesicle-derived mRNA reflected the treatment effect and provided implications of mRNA expression in cancer cells (**Figure 3**).
- In the blood of breast cancer patients, the *HER2* level detected in EVs reflected the *HER2* status of the primary tumor (**Figure 3D**).

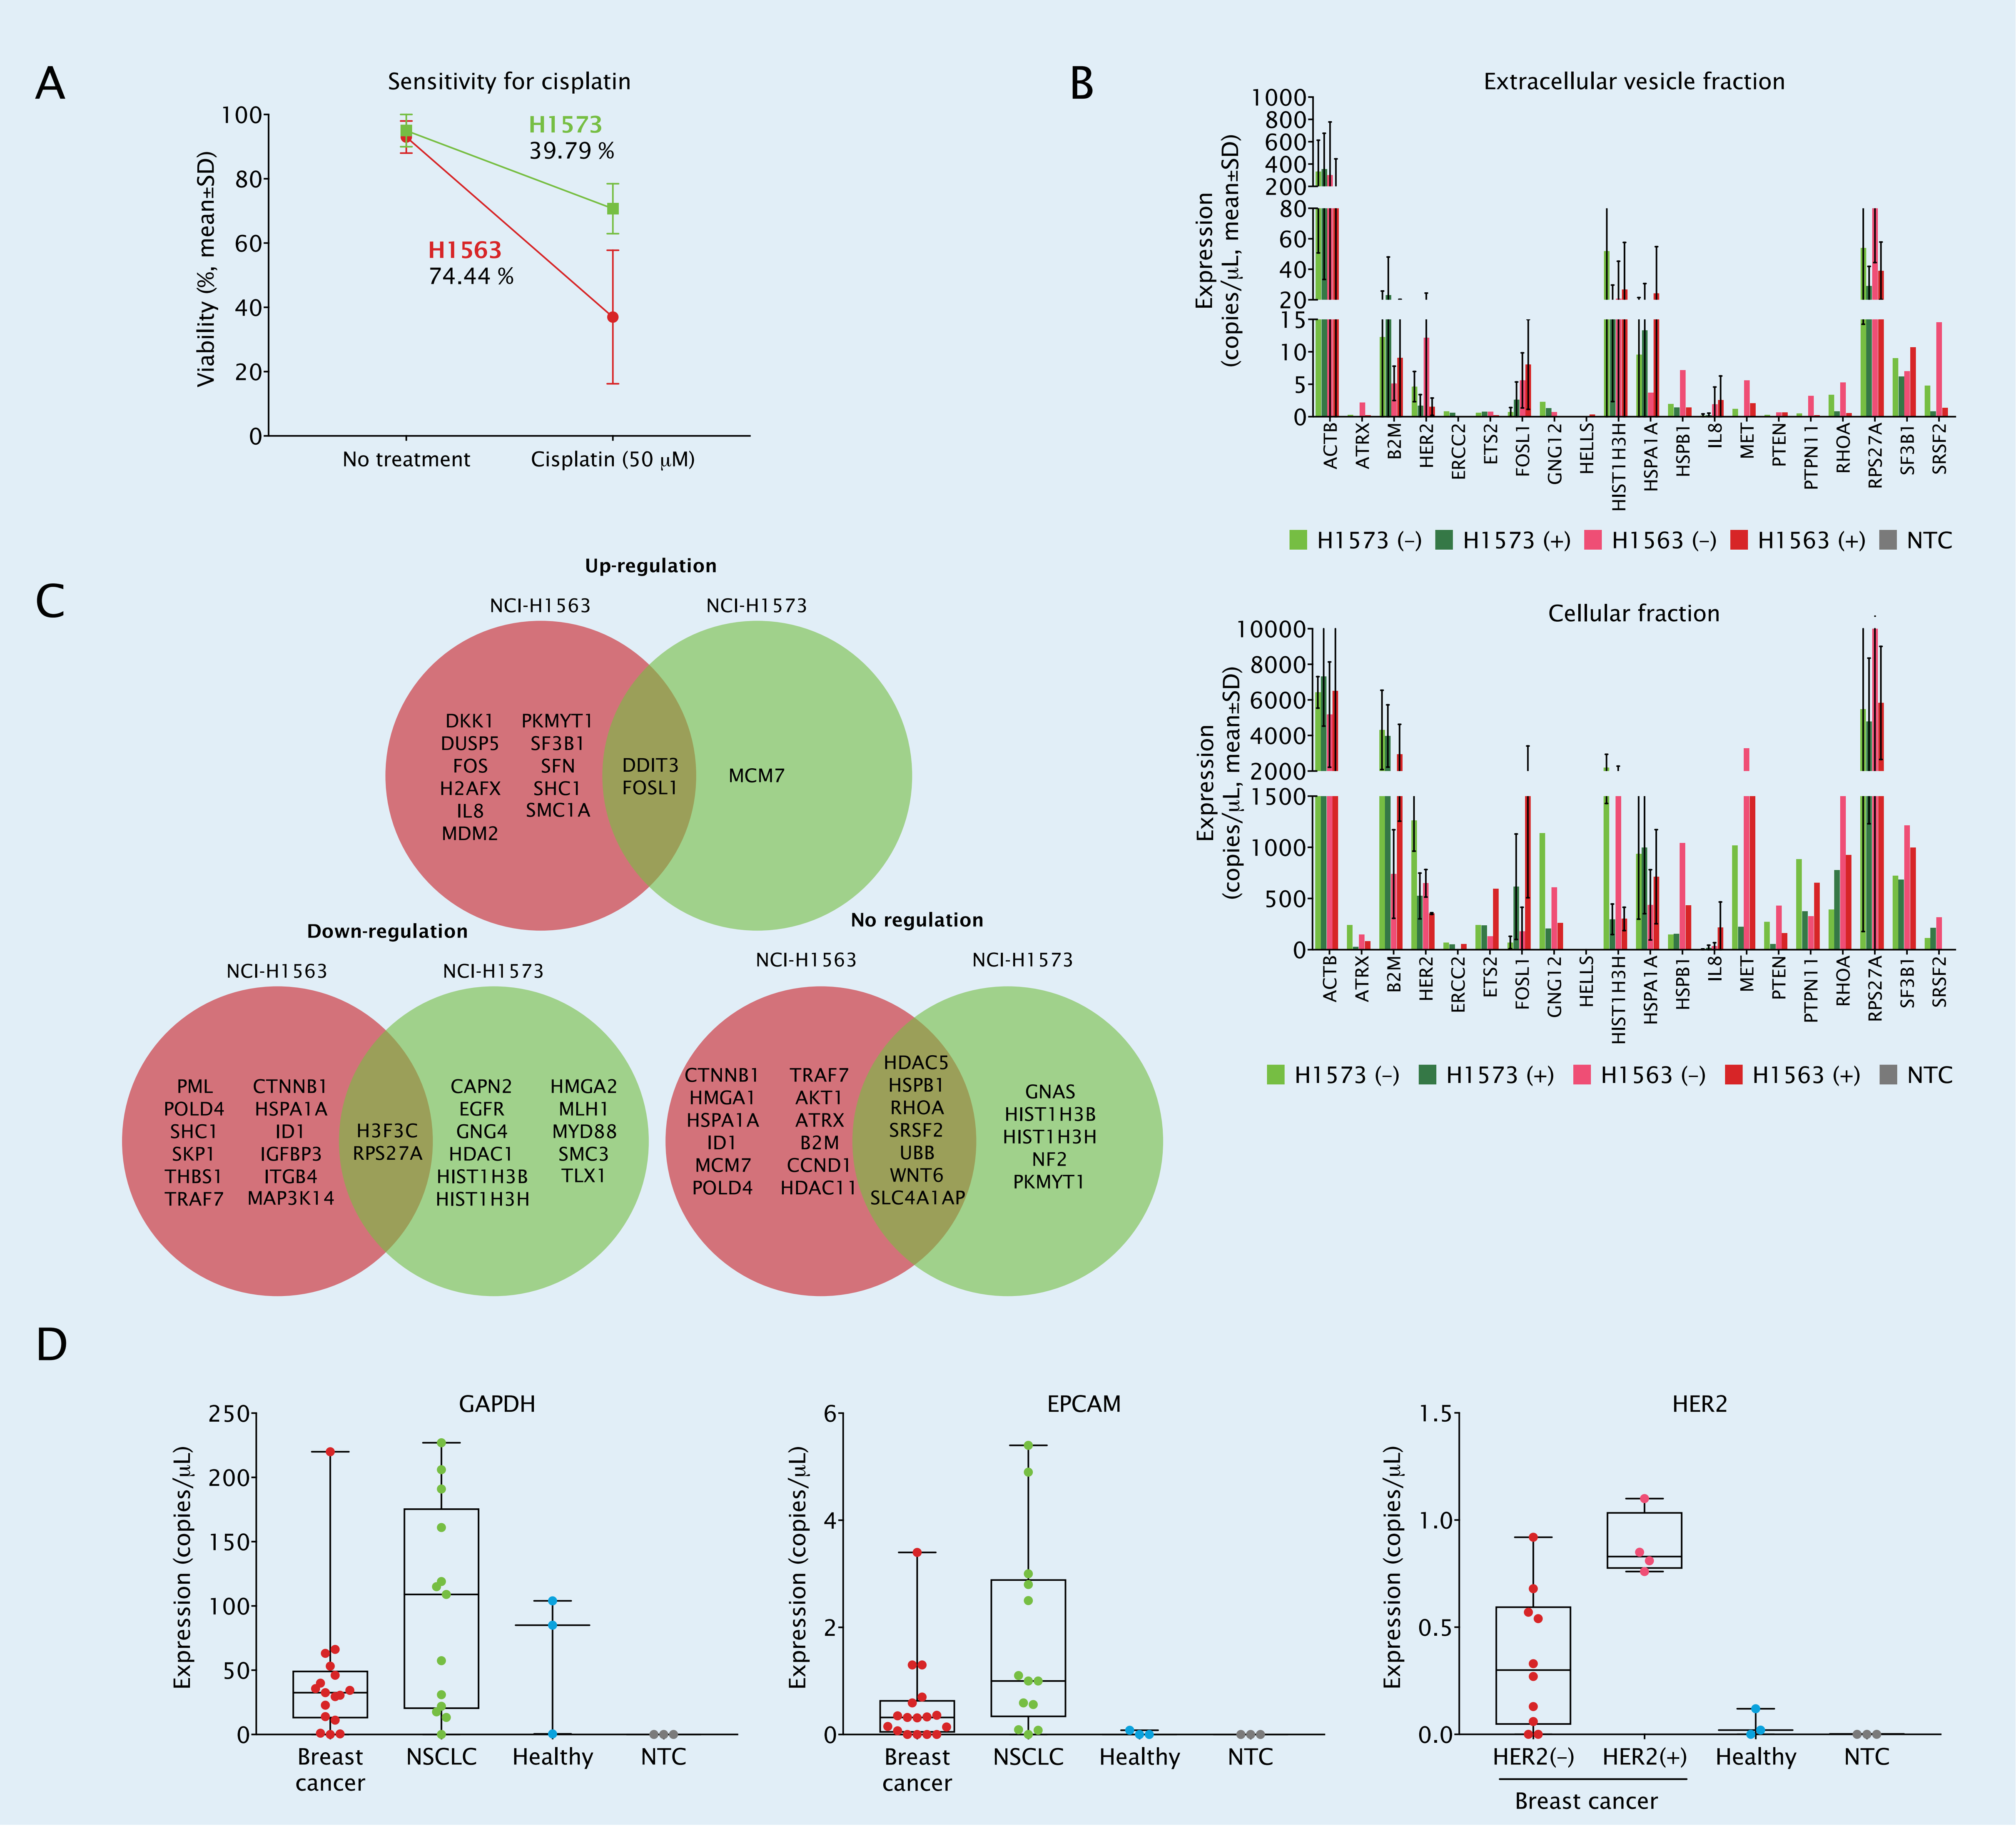
A
Sensitivity for cisplatin
Viability (%, mean±SD)
H1573
39.79 %
H1563
74.44 %
No treatment
Cisplatin (50 µM)
B
Extracellular vesicle fraction
Cellular fraction
Expression (copies/µL, mean±SD)
ACTB
ATRX
B2M
HER2
ERCC2
ETS2
FOSL1
GNG12
HELLS
HIST1H3H
HSPA1A
HSPB1
IL8
MET
PTEN
PTPN11
RHOA
RPS27A
SF3B1
SRSF2
H1573 (–)
H1573 (+)
H1563 (–)
H1563 (+)
NTC
C
Up-regulation
NCI-H1563
NCI-H1573
DKK1
DUSP5
FOS
H2AFX
IL8
MDM2
PKMYT1
SF3B1
SFN
SHC1
SMC1A
DDIT3
FOSL1
MCM7
Down-regulation
PML
POLD4
SHC1
SKP1
THBS1
TRAF7
CTNNB1
HSPA1A
ID1
IGFBP3
ITGB4
MAP3K14
H3F3C
RPS27A
CAPN2
EGFR
GNG4
HDAC1
HIST1H3B
HIST1H3H
HMGA2
MLH1
MYD88
SMC3
TLX1
No regulation
CTNNB1
HMGA1
HSPA1A
ID1
MCM7
POLD4
TRAF7
AKT1
ATRX
B2M
CCND1
HDAC11
HDAC5
HSPB1
RHOA
SRSF2
UBB
WNT6
SLC4A1AP
GNAS
HIST1H3B
HIST1H3H
NF2
PKMYT1
D
GAPDH
EPCAM
HER2
Expression (copies/µL)
Breast cancer
NSCLC
Healthy
NTC
HER2(–)
HER2(+)

Figure 3. Detection of extracellular vesicle-derived mRNA (EVmRNA) in cancer cells. H1563 and NCI-H1573 (H1573) NSCLC cells were treated with 50 µM cisplatin for 24 h. The supernatant and cellular fractions were collected and EVmRNA extraction was performed.

A) H1573 cells were found to be more resistant to cisplatin treatment than H1563 cells (percentage of dead cells 39.79 and 77.44%, respectively, compared to a non-treated control).

B) Changes in gene expression in H1563 and H1573 cells (EVs or cellular fraction) with (+) or without (-) cisplatin treatment as detected by ddPCR. Altered mRNA expression in response to cisplatin treatment was observed for example for *FOSL1* both in the EVs and the cellular fraction. *HER2* and *RPS27A* showed downregulation upon treatment. No changes were observed for *ACTB*. Interestingly, *HIST1H3H* showed significant changes in the cellular fraction, but not in the EV fraction. The experiments were performed with three independent biological replicates.

C) To determine if a multiplex target quantification is possible, a PanCancer Immune Profiling assay (NanoString) was performed. Only target genes which were 1.5-fold above background and showing a 2-fold differential expression compared to the control are shown. Some genes were up- or downregulated (e.g. up: *FOSL1*, down: *RPS27A*) or significantly above the background (e.g. *H2PB1*) in both cell lines. The expression of *FOSL1* and *RPS12A* was validated by ddPCR.

D) EVmRNA profiling of *GAPDH*, *EpCAM* and *HER2* in plasma samples from patients with breast cancer or NSCLC, as well as from healthy volunteers (n=3). The *HER2* expression level in the EV fraction reflected the *HER2*-negative or positive primary tumor status in breast cancer patients. High expression of *EpCAM* or *HER2* was observed specifically in cancer specimens while those targets were not detected in plasma samples from healthy donors.